Life without MSCs :
Determining the effects of stromal cell loss

Camarillo E.[1], Duffy M.[1], O'Flynn L.[1], Watson L. [1], Bahri O[2]., McCullagh K[2]., Thompson K.[3], Canney M.[3], Dockery P.[3], Ceredig R.[2], Elliman S.J.[1]

[1]Orbsen Therapeutics Ltd, [2]Regenerative Medicine Institute, [3]Department of Anatomy, NUI Galway, University Rd., Ireland eva.camarillo@orbsentherapeutics.com

***Abstract*:** Mesenchymal stromal cells (MSCs) are considered to be promising agents for the treatment of immunological diseases. Although originally identified as precursor cells for mesenchymal lineages, *in vitro* studies and phase 1 and 2 clinical trials have demonstrated immune-modulatory effects after MSC infusion. Nevertheless, the endogenous identity, function and heterogeneity of MSC are only beginning to be elucidated. Fearon et al. (2010, 2012) identified Fibroblast Activation Arotein-α (FAP) as a marker of adult-tissue resident MSC. To assess the role of FAP⁺MSC *Kraman et al.* (2010) created a transgenic (Tg) mouse with Bacterial Artificial Chromosomes (BACs) containing the murine *fap* gene modified by insertion of a cassette encoding the primate Diphtheria Toxin Receptor (DTR), Luciferase (Luc2) and the fluorescent reporter mCherry (Figure 1B) and designating the strain, FAP^{DM2}. FAP⁺MSC can be specifically deleted in FAP^{DM2} mice using a simple and sensitive Diptheria Toxin (DTX) receptor-mediated conditional cell knockout (TRECK) (Figure 1A). Fearon and colleagues (2012) reported for the first time that DTX-mediated ablation of FAP⁺ cells led to a marrow anemia, perturbed hematopoiesis and muscle cachexia.

Orbsen Therapeutics have established the only reported MSC knockout mouse (FAP^{DM2}) and optimised FAP⁺ cell albation protocols to investigate:

1) Are the three reported phenotypes associated with FAP⁺cell ablation reproducible?
2) Can these phenotypes associated with stromal cell loss be rescued using stromal cell therapy?
3) More broadly, do the phenotypes associated with stromal cell loss identify "stromal cell diseases" that could be appropriately treated with stromal cell therapy?

Methods: FAP⁺cell ablation is achieved by administering the FAP-Tg mouse with 2x intraperitoneal (I.P.) injections of Diphtheria Toxin (DTX – 25ng/g body weight) on Day 1 and Day 3, followed by monitoring of the animal for 72hrs. FAP⁺cell ablation was confirmed by flow cytometry at 72hrs after the final dose of DTX (Figure 2B). Body weight was monitored for 10 days pre-injection and at the end point 72 hrs (Figure 3A). Blood was collected by intracardiac puncture for haemoglobin measurement and centrifuged for serum biochemistry analysis (Figure 3B and C). Bone Marrow (BM) from femur and tibia were harvested and digested by an enzymatic cocktail and stained for MSC (CD362 and FAP) (Figure 2B) and Hematopoietic stem cell (HSC) surface markers (Lineage, c-kit, Sca-1, CD34, CD48, CD135, CD150, and IL-7R) (Figure 4). Flow cytometry was carried out on the FACS Canto II. Sternum BM was collected, decalcified softly with an acidic agent, frozen and sectioned at 5 microns in the cryostat for histological analysis by using haematoxylin and eosin (H&E) staining (Figure 3E). Statistical analyses were conducted using a one-way Anova test and Tukey's multiple comparisons test, unless otherwise specified; *p<0.05, ***p<0.001, ****p<0.0001.

Results: Fearon and colleagues (2012) reported that FAP⁺cell ablation leads to anemia, altered hematopoiesis and cachexia. We have independently confirmed the findings that DTX-treatment of FAP-Tg mice causes:

(1) FAP⁺cell ablation; (2) weight loss; (3) anemia (low Hb and low ALP) and (4) perturbed hematopoiesis In addition, we report that FAP⁺cell ablation leads to marrow hemorrhaging, followed by early stage fibrosis of the marrow cavity by ECM deposition. Preliminary data indicates that stromal cell therapy alleviates ablation-induced marrow fibrosis.

Toxin receptor-mediated conditional cell knockout (TRECK)

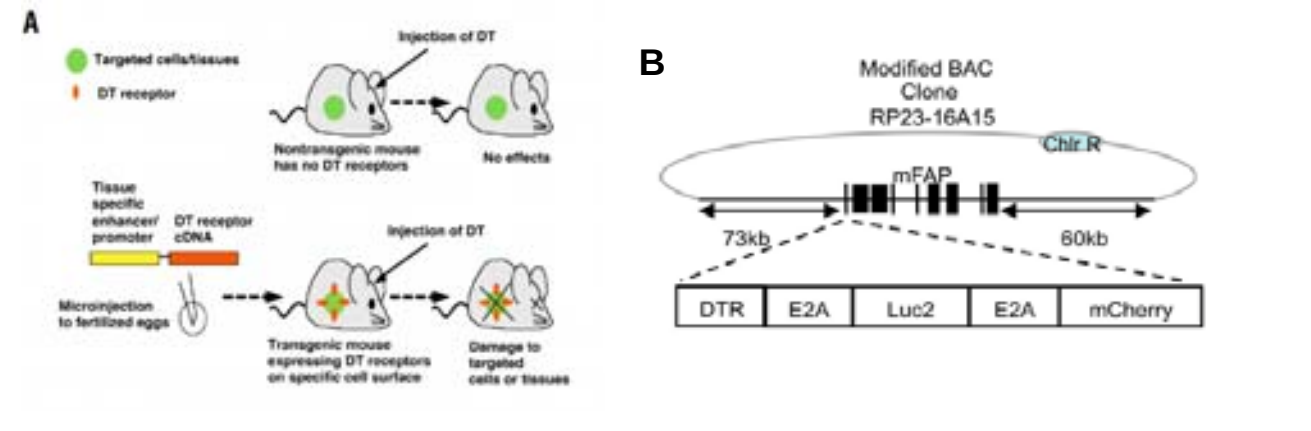
A
Targeted cells/tissues
DT receptor
Injection of DT
Nontransgenic mouse has no DT receptors
No effects
Tissue specific enhancer/ promoter
DT receptor cDNA
Microinjection to fertilized eggs
Transgenic mouse expressing DT receptors on specific cell surface
Damage to targeted cells or tissues
B
Modified BAC Clone RP23-16A15
Chlr R
mFAP
73kb
60kb
DTR
E2A
Luc2
E2A
mCherry

Figure 1. Animal model: (A) Toxin receptor-mediated conditional cell knockout (TRECK) procedure from Saito M. Nature 2001 .(B) The *fap* BAC transgene construction from Roberts EW. J Exp Med. 2013

Intraperitoneal injection of DTX causes FAP⁺ SSC ablation

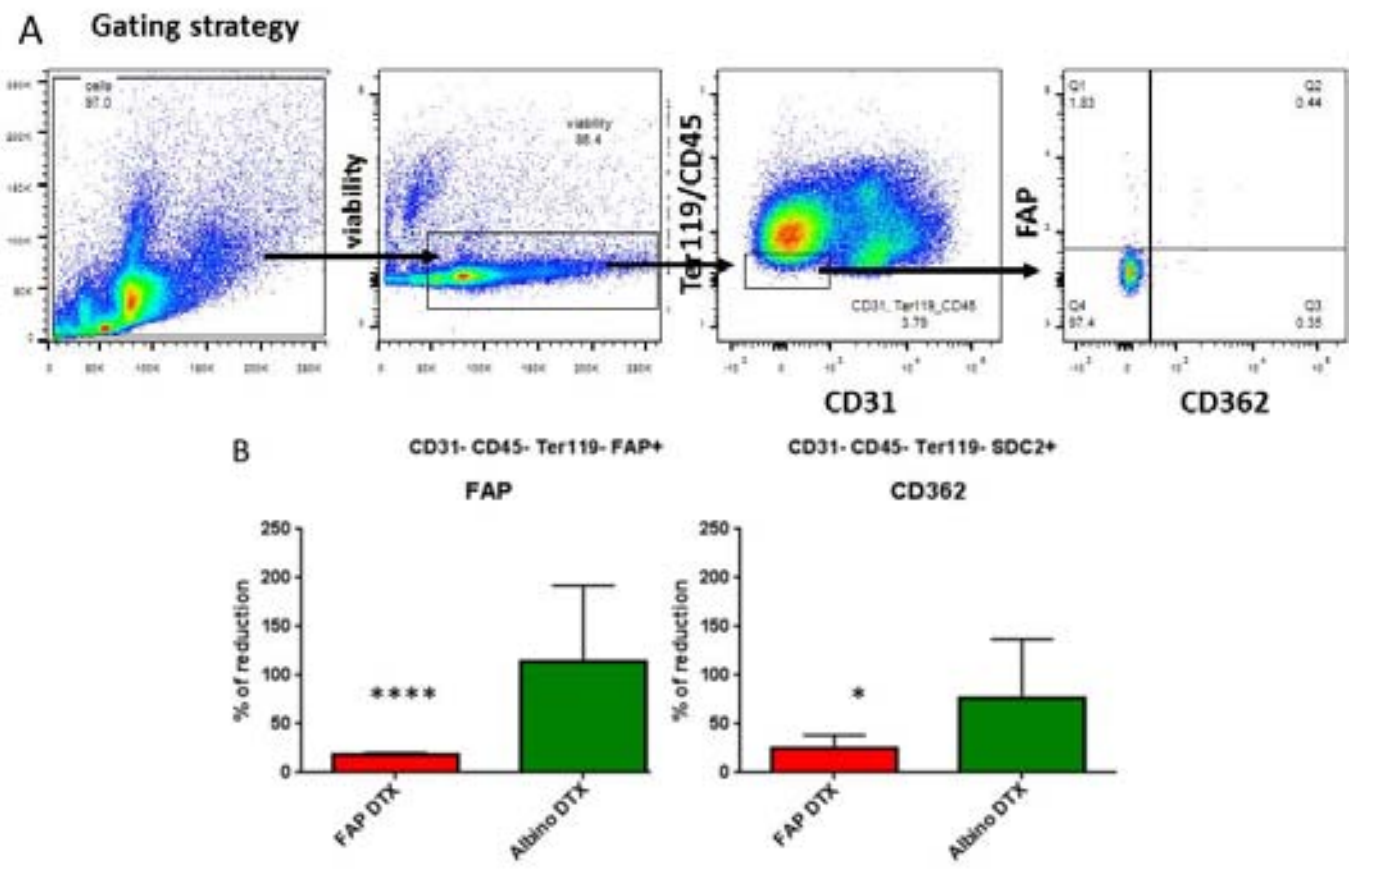
A Gating strategy
viability
Ter119/CD45
FAP
CD31
CD362
B
CD31- CD45- Ter119- FAP+
FAP
CD31- CD45- Ter119- SDC2+
CD362
% of reduction

*
FAP DTX
Albino DTX

Figure 2. FAP⁺ stromal cell ablation. (A) The expression of FAP⁺ and CD362⁺ cells on Ter119⁻, CD45⁻, CD31⁻ cell population suspensions from the BM was determined by flow cytometry. (B) Bar graphs indicate siginificant reduction in the percentage of FAP⁺ stromal cells after DTX injection in FAP group. Moreover, the percentage of CD362 is also significant y reduced after FAP⁺ MSC ablation.

FAP⁺cell ablation causes Anaemia and Hemorrhaging

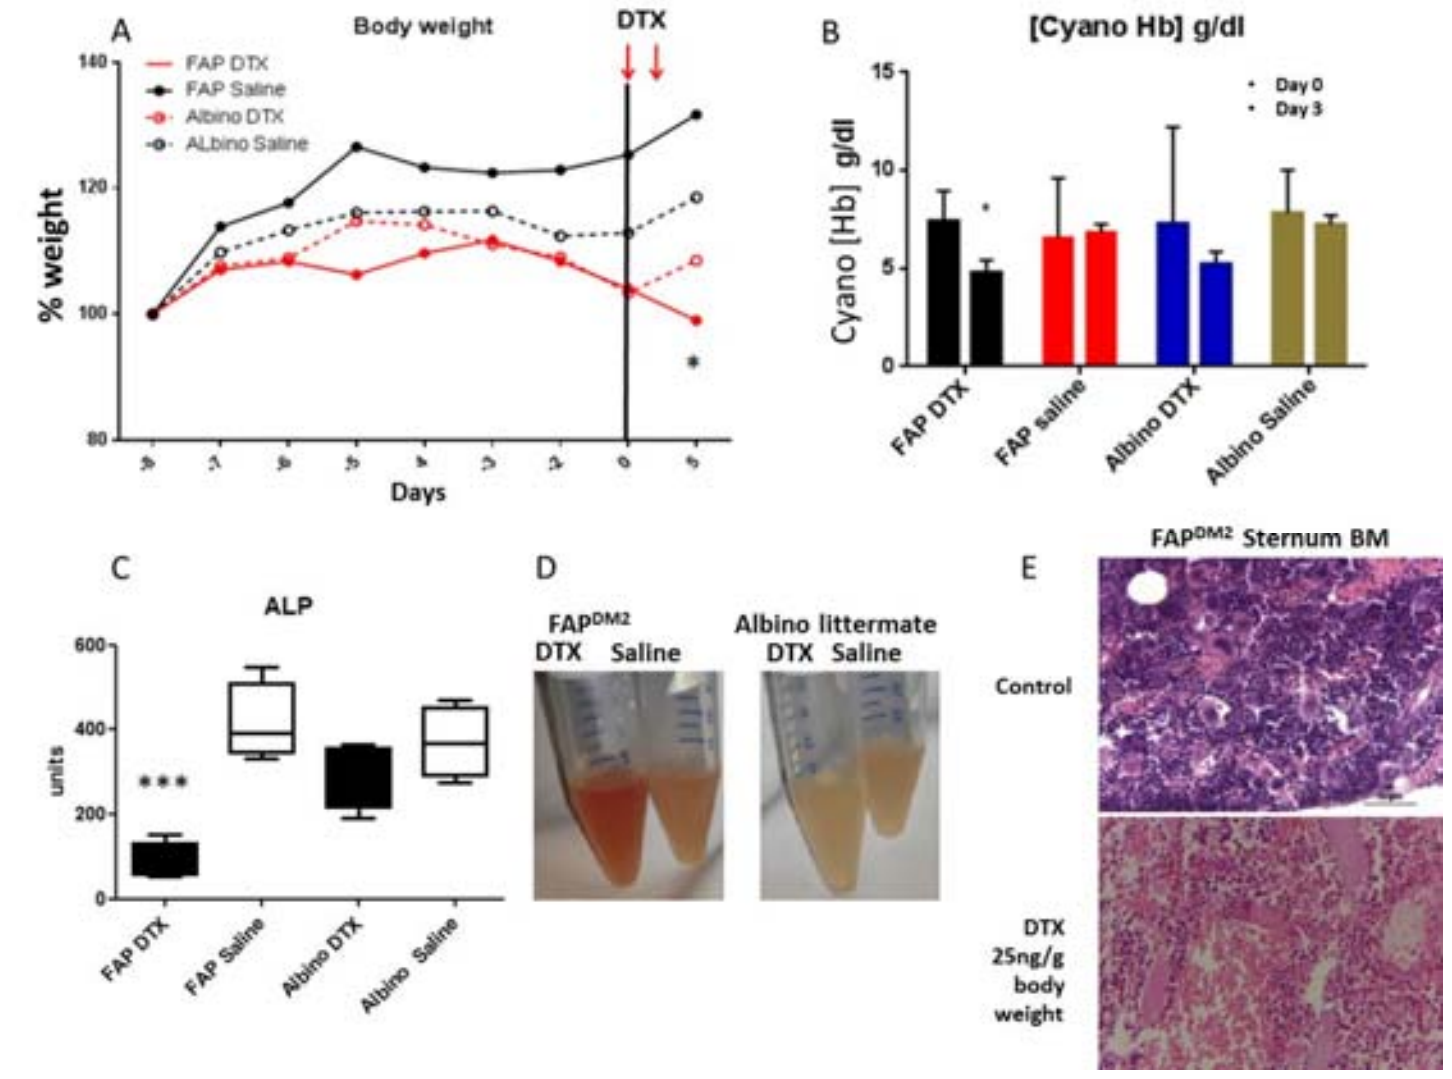
A
Body weight
DTX
FAP DTX
FAP Saline
Albino DTX
ALbino Saline
% weight
Days
B
[Cyano Hb] g/dl
Day 0
Day 3
Cyano [Hb] g/dl
FAP DTX
FAP saline
Albino DTX
Albino Saline
C
ALP
units

FAP DTX
FAP Saline
Albino DTX
Albino Saline
D
FAPDM2
DTX Saline
Albino littermate
DTX Saline
E
FAPDM2 Sternum BM
Control
DTX 25ng/g body weight

Figure 3. Anaemia phenotype: (A) the graph shows the statistical y significant decrease in total body weight at the end point in the DTX treated FAP^{DM2} group. (B) [Hb] measurement at day 0 is compared to the end point day 3 (72 hours after the second boost DTX) demonstrating a statistical significant decrease in the DTX treated FAP^{DM2} group. (C) ALP in serum decreased in DTX treated FAP^{DM2} group. (D) Cell suspension from digested bone marrow (BM) before flow cytometry staining showing increased numbers of erythrocytes (red color) in the FAP^{DM2} DTX group. (E) H&E sections at 20X show the bleeding episode in the DTX treated FAP^{DM2} group at 72 hours after the second boost of DTX administration.

FAP⁺ cell ablation alters murine haematopoiesis

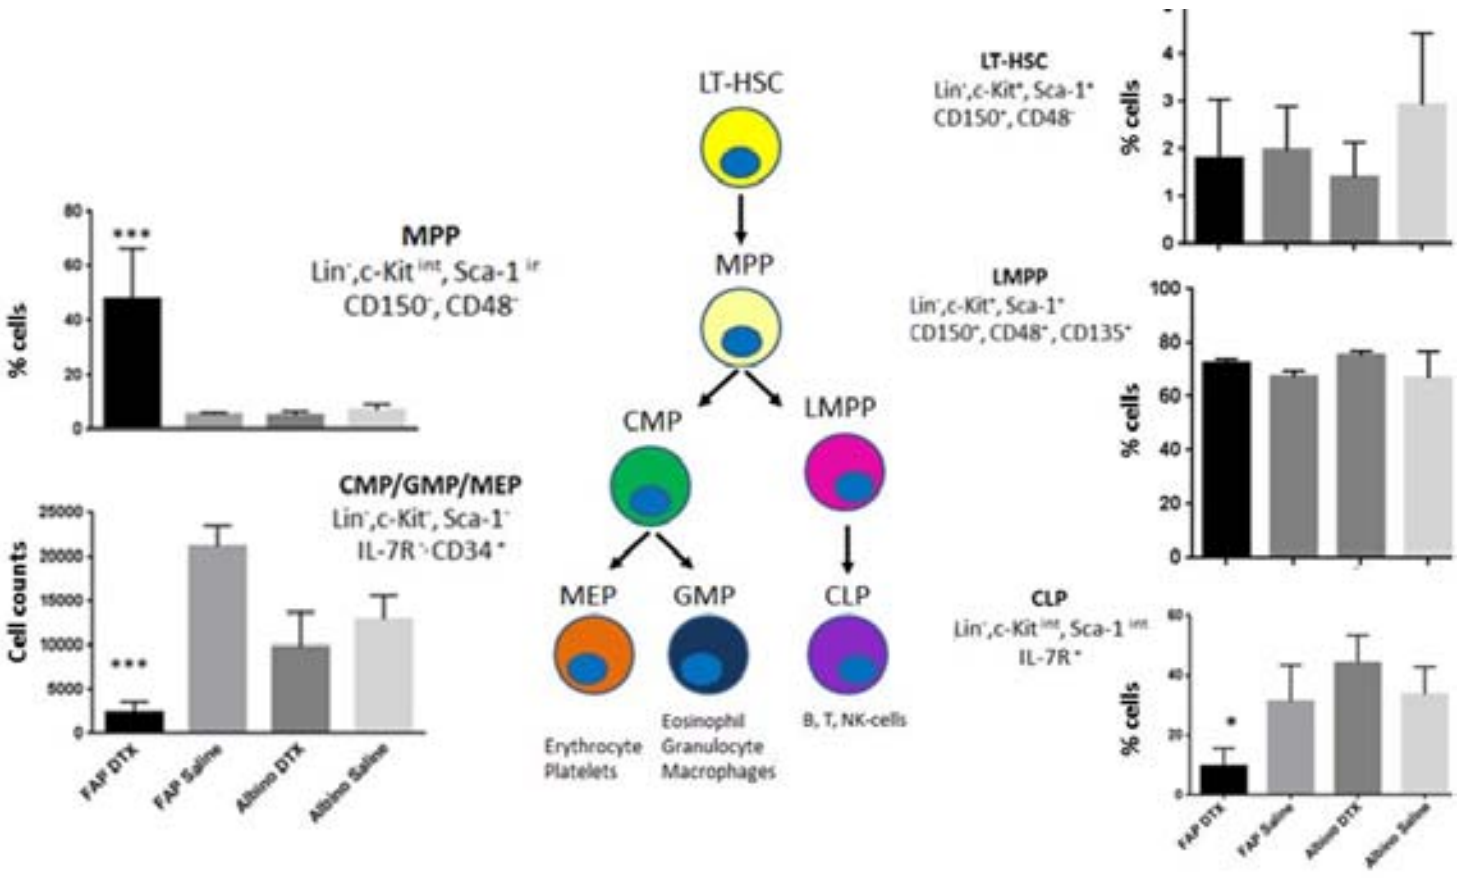
LT-HSC
MPP
CMP
LMPP
MEP
GMP
CLP
Erythrocyte Platelets
Eosinophil Granulocyte Macrophages
B, T, NK-cells
MPP
CMP/GMP/MEP
Cell counts
% cells

*
FAP DTX
FAP Saline
Albino DTX
Albino Saline

Figure 4. FAP⁺cell ablation peturbs BM hematopoiesis: (A) An illustration of the Hematopoietic hierarchy for the generation of LMPP (lymphoid-primed multipotent progenitor), CLP (common lymphoid progenitor), CMP (common myeloid progenitor) for GMP (granulocyte myeloid progenitor), and MEP (Megakaryocyte progenitors) adapted from *Grover et al 2014*. FAP stromal cell ablation has shown an altered haematopoiesis with a statistically significant increased percentage of MPP, reduced percentage of CLP and reduced cells counts in CMP/GMP/MEP progenitors for FAP DTX group.

Can Stromal stem cell administration rescue FAP ablation?

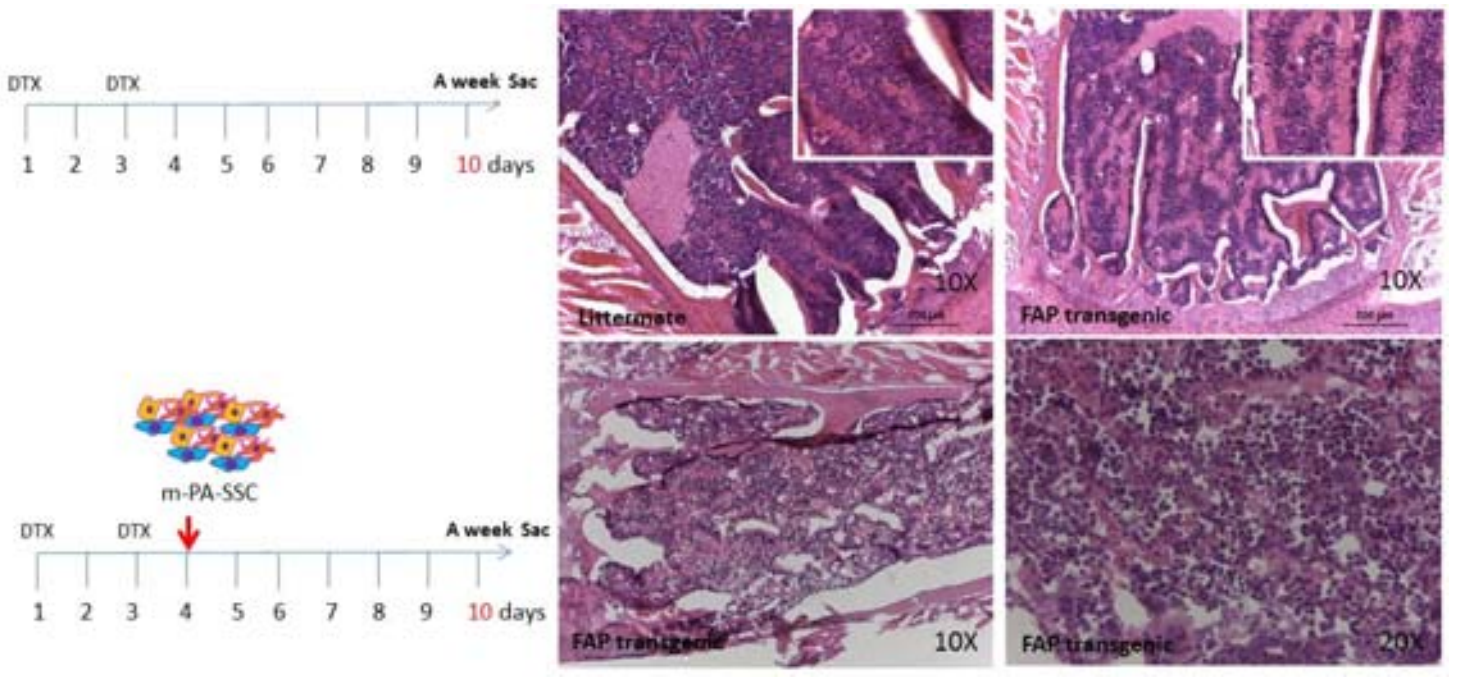
DTX
DTX
A week Sac
1 2 3 4 5 6 7 8 9 10 days
m-PA-SSC
Littermate
FAP transgenic
10X
20X

Figure 5. Stromal cell therapy rescues early Fibrosis phase in bone marrow cavity. Top row images show H&E staining of sternum bone cavity indicating ECM in pink one week after the final dose of DTX in a representative FAP transgenic animal. The image is compared to a representative littermate animal. Bottom row images show the effect of a mouse SSC cell administration 24hrs after final dose of DTX . ECM deposition is reduced at day 7.

Conclusions

1. We confirm that FAP⁺ cell ablation causes anaemia and altered haematopoiesis in the FAP^{DM2} model.
2. We report for the first time FAP⁺ cell ablation causes haemorrhaging and fibrosis. Preliminary data indicates stromal cell therapy can rescue this fibrosis.
3. Ongoing studies will address if the cachectic pheotype reported in the FAP^{DM2} model is reproducible.
4. Further experiments will assess if stromal cell therapy can rescue the anemia, peturbed haematopoisis (and cachexia) associated with FAP⁺cell ablation.

References

1. Grover, A., et al., Erythropoietin guides multipotent hematopoietic progenitor cells toward an erythroid fate. J Exp Med, 2014. 211(2): p. 181-188.
2. 2Kraman, M., et al., Suppression of antitumor immunity by stromal cells expressing fibroblast activation protein-alpha. Science, 2010. 330(6005): p. 827-30.
3. Roberts, E.W., et al., Depletion of stromal cells expressing fibroblast activation protein-alpha from skeletal muscle and bone marrow results in cachexia and anaemia. J Exp Med, 2013. 210(6): p. 1137-51.

DECIDE

Marie Sklodowska-Curie Initial Training Networks

Grant No. EU FP7/315902

European Commission Seventh Framework Programme

NUI Galway OÉ Gaillimh